USING CAUSAL AI *IN-SILICO* PATIENT MODEL TO INFER CANCER CELL GENE DEPENDENCY IN MULTIPLE MYELOMA

American Association for Cancer Research®

Abstract Number: LB181

BRANDON NATHASINGH[1], DEREK WALKAMA[1], LAUREL MAYHEW[1], KENDALL LOH[1], JEANNE LATOURELLE[1], BRUCE CHURCH[1], YAOYU E. WANG[1]

1. Aitia, Somerville, MA

Contact: ywang@aitiabio.com

SUMMARY

Causal inference network models are particularly suited to facilitate therapeutic intervention studies by inferring the molecular causal structure from multimodal data. Here, we developed a multiple myeloma (MM) Digital Twin model of overall survival (OS) to perform full transcriptome *in-silico* loss-of-function experiments. We compared the results to MM cell lines CRISPR knock-out data from DepMap[1] to show high concordance. The model also identifies additional dependent genes and provides potential mechanisms of action. This demonstrates *in-silico* AI causal modelling is a powerful tool for exploring cancer cell vulnerability from patient data to advance target discovery.

AIMS

We aim to demonstrate that our multiple myeloma Digital Twin model can identify cancer cell dependent genes with *in-silico* knockdown experiments, identify response biomarkers, and suggest mechanisms of action.

RESULTS

The *in-silico* loss-of-function experiments on the MM Digital Twin model identified 102 OS driver genes that causally drive patient overall survival. Most of the coding genes are found to be MM-Dependent in DepMap. **PHF19**, a gene known to control MM cell proliferation[2], was found to be non-dependent in the DepMap but was identified as an OS driver by our model.

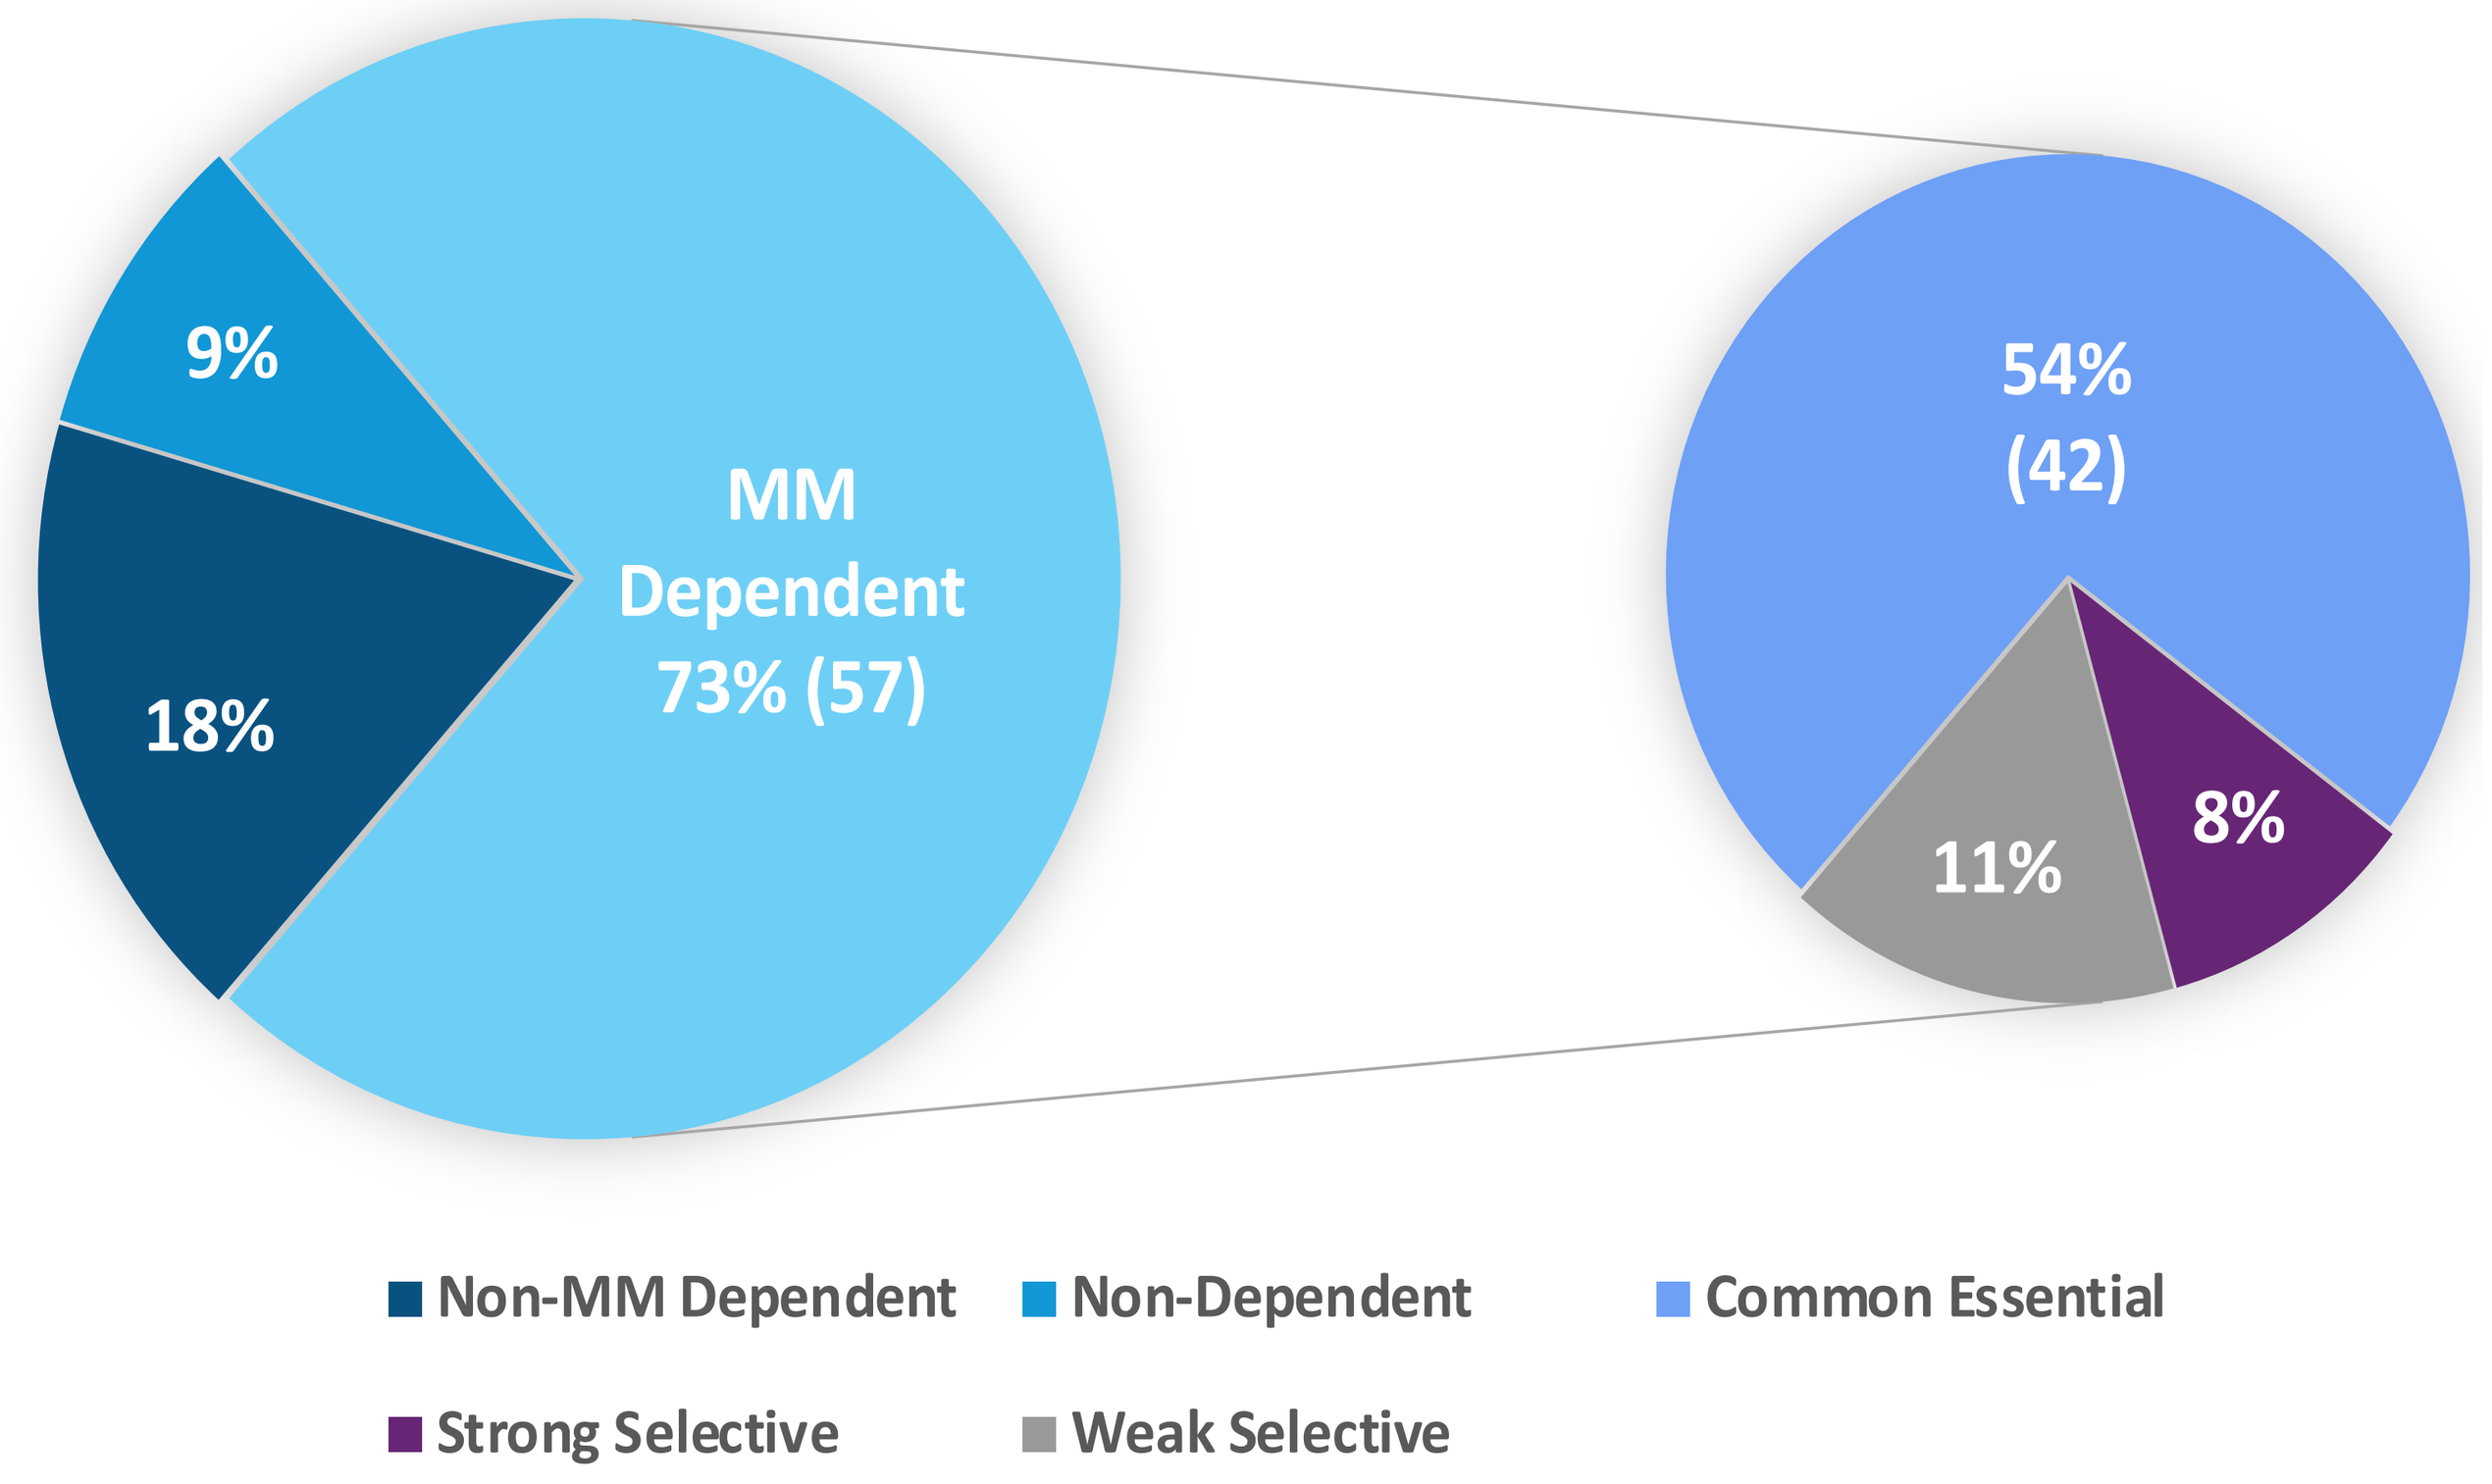

9%
18%
MM Dependent 73% (57)
54% (42)
11%
8%
Non-MM Dependent
Non-Dependent
Common Essential
Strong Selective
Weak Selective

Figure 2: DepMap Annotation of OS Driver Genes. 73% (57/78, p<2.2e-16, OR=9.6) of the coding genes are dependent in MM cell lines, with 42 common essential, 6 strong selective and 9 weak selective genes. The remaining 21 OS driver genes, 14 (66%) are non-MM Dependent.

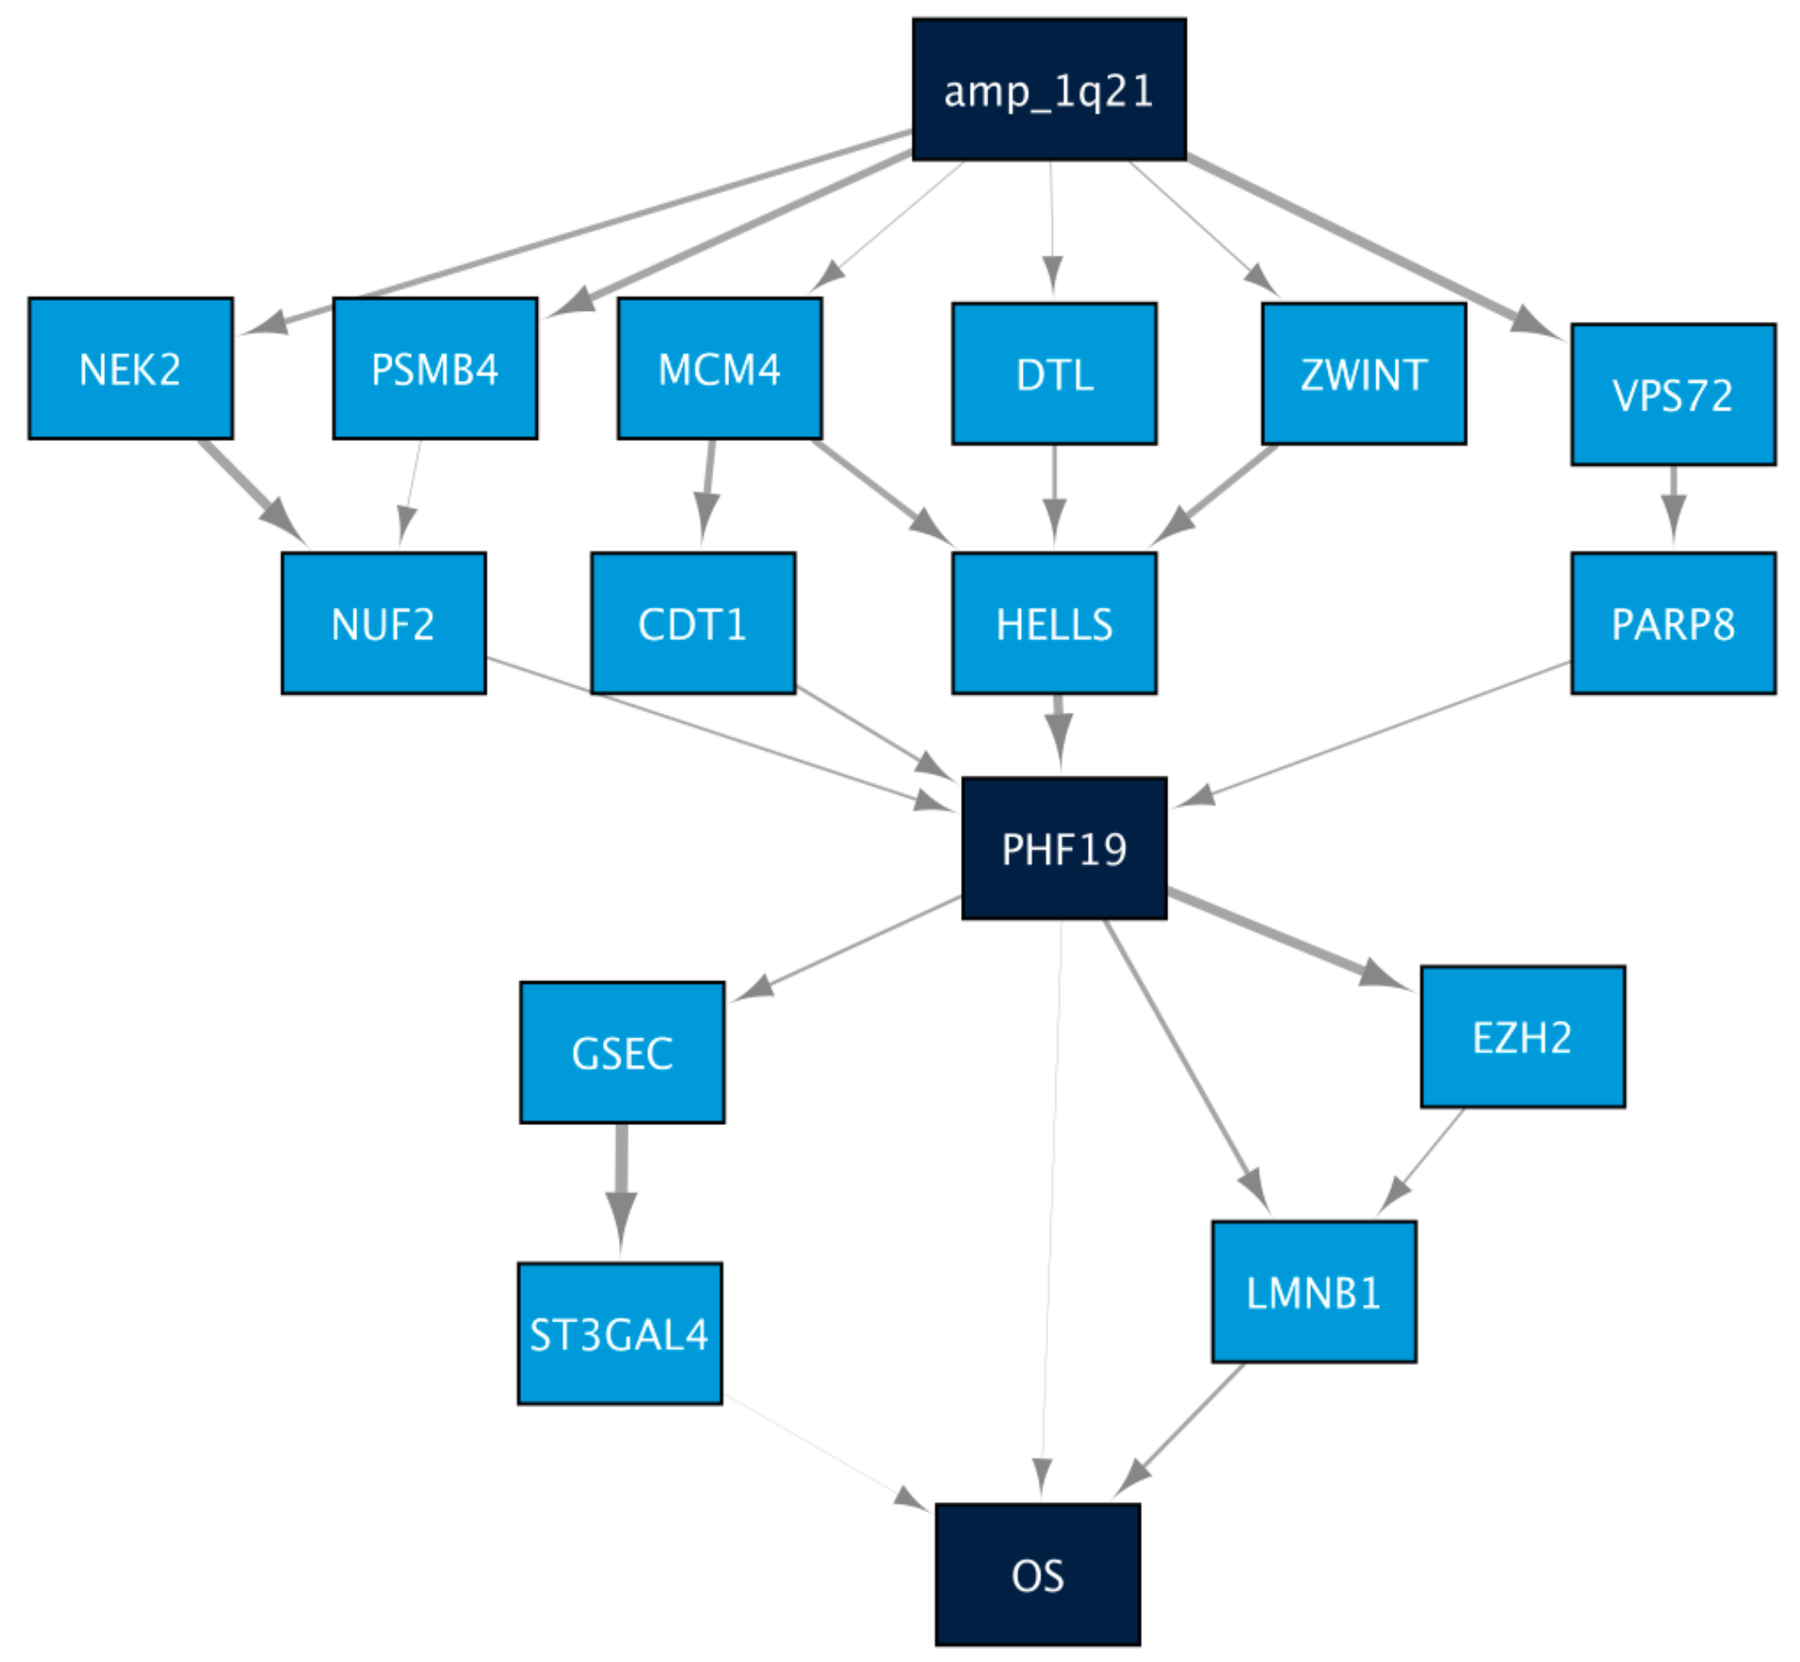

amp_1q21
NEK2
PSMB4
MCM4
DTL
ZWINT
VPS72
NUF2
CDT1
HELLS
PARP8
PHF19
GSEC
EZH2
ST3GAL4
LMNB1
OS

Figure 3: Model Topology around PHF19: The PHF19 local network suggests 1q21 amplification causally affects PHF19 and patient OS.

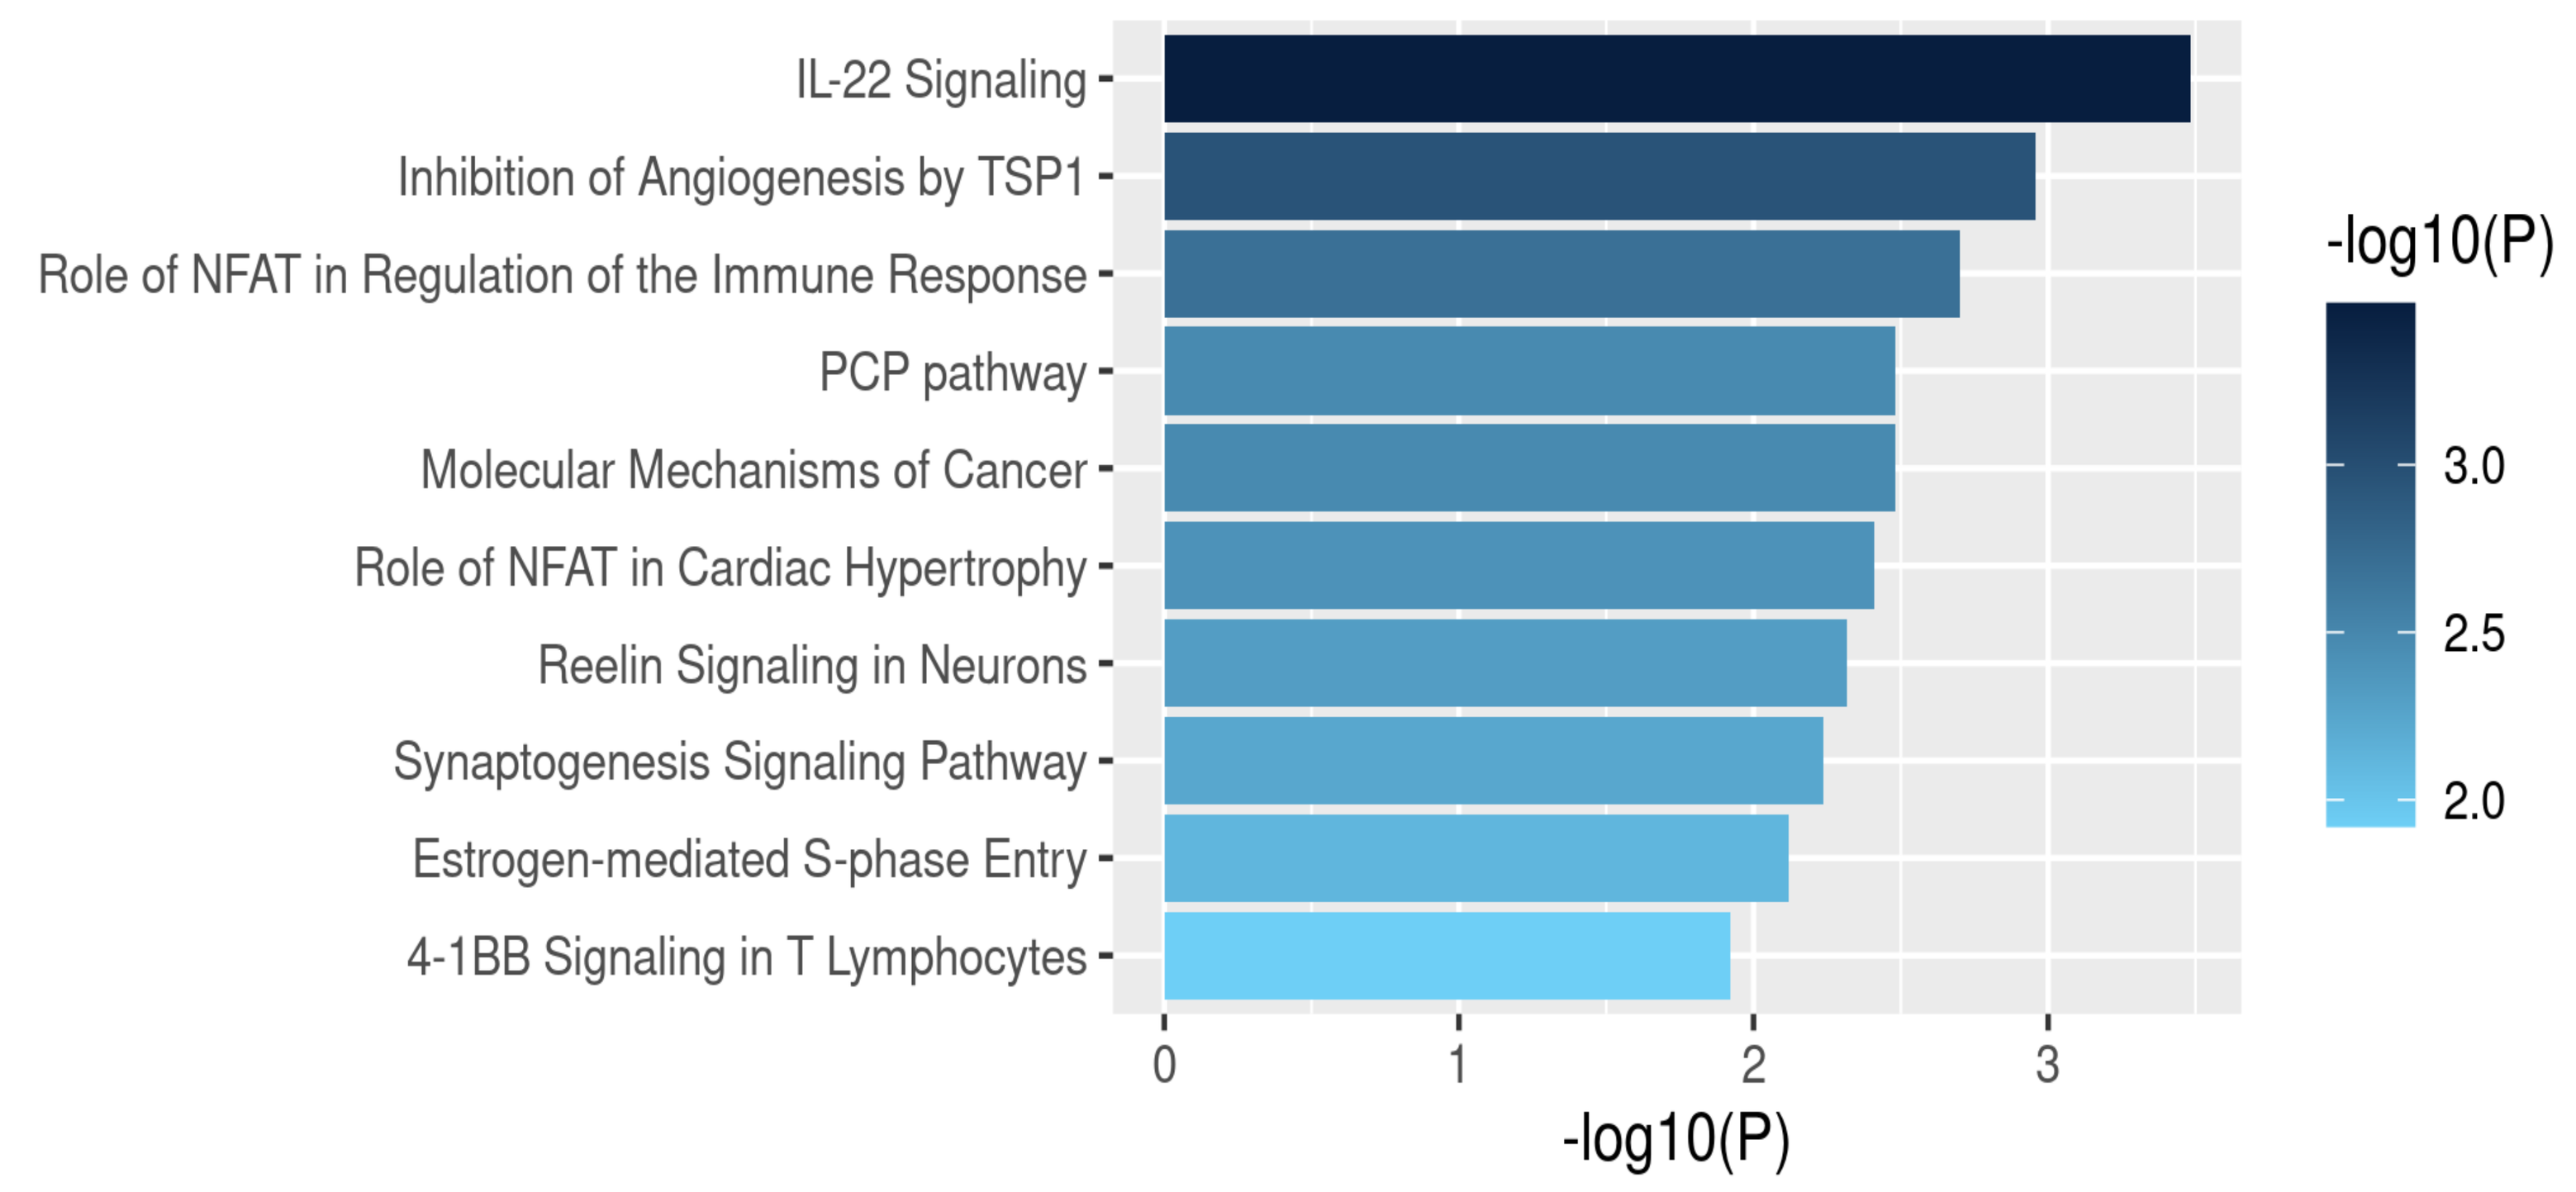

IL-22 Signaling
Inhibition of Angiogenesis by TSP1
Role of NFAT in Regulation of the Immune Response
PCP pathway
Molecular Mechanisms of Cancer
Role of NFAT in Cardiac Hypertrophy
Reelin Signaling in Neurons
Synaptogenesis Signaling Pathway
Estrogen-mediated S-phase Entry
4-1BB Signaling in T Lymphocytes
-log10(P)
3.0
2.5
2.0
0
1
2
3
-log10(P)

Figure 5: Biological pathways driven by PHF19: 162 downstream genes are enriched in report processes in cell proliferation and angiogenesis[3].

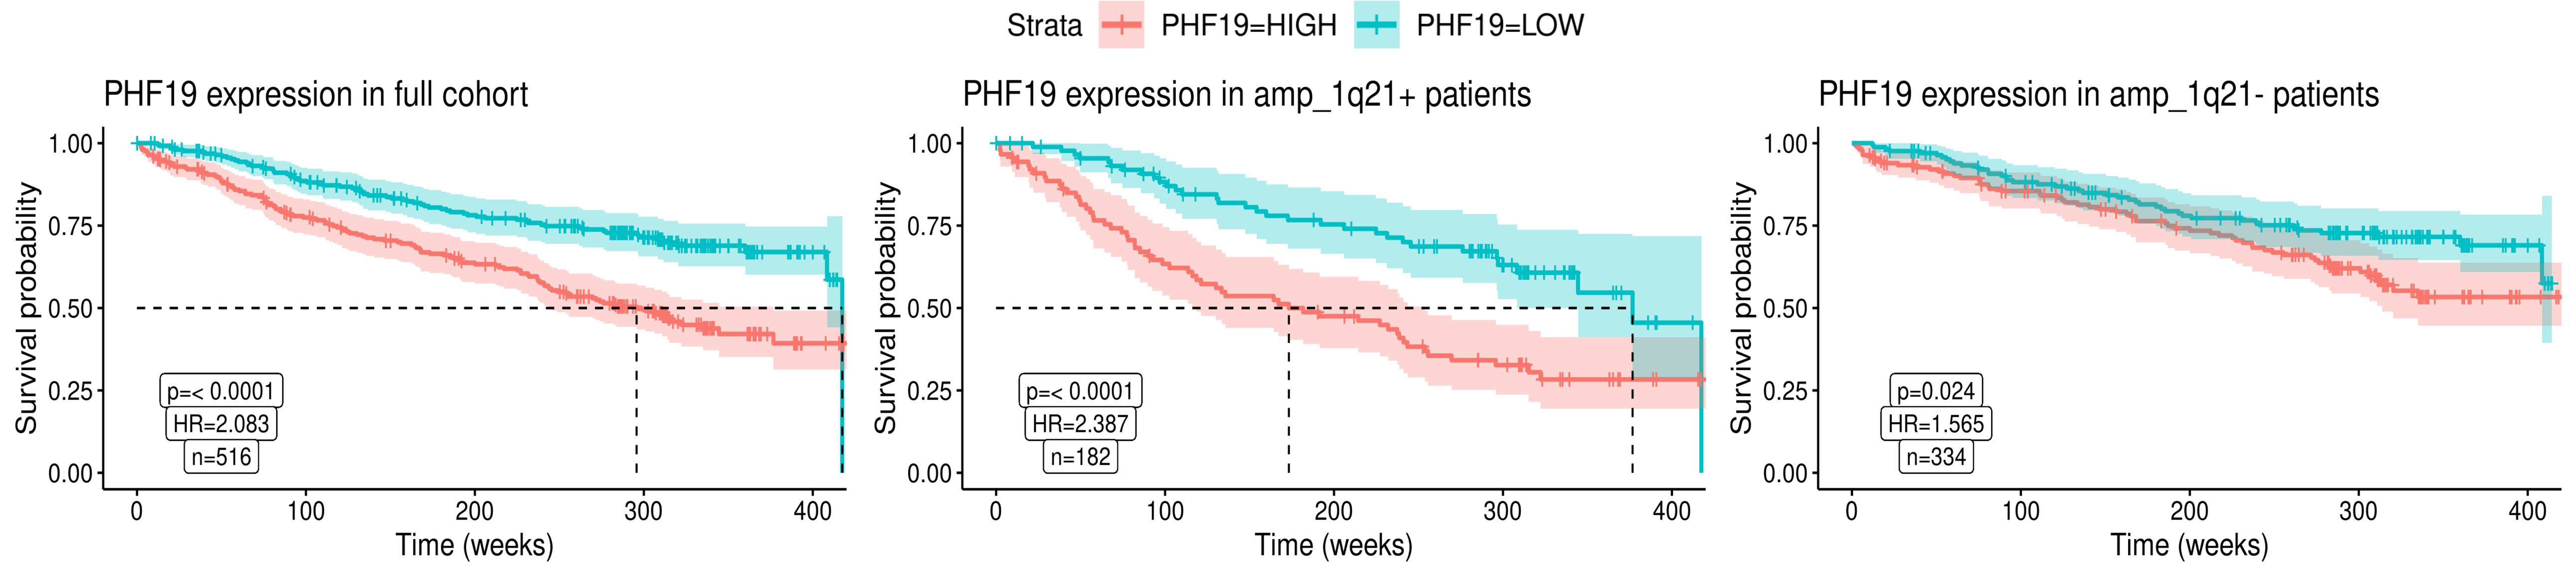

Strata PHF19=HIGH PHF19=LOW
PHF19 expression in full cohort
PHF19 expression in amp_1q21+ patients
PHF19 expression in amp_1q21- patients
Survival probability
1.00
0.75
0.50
0.25
0.00
p=< 0.0001
HR=2.083
n=516
p=< 0.0001
HR=2.387
n=182
p=0.024
HR=1.565
n=334
0
100
200
300
400
Time (weeks)

Figure 4: Response biomarkers for PHF19: Kaplan Meier plots for conditioned subpopulations stratified by median PHF19 expression.

METHODS

We constructed an *in-silico* multiple myeloma (MM) patient causal model of overall survival (OS) based on transcriptomic expression, clinical, and genomic alteration data from Multiple Myeloma Research Foundation (MMRF) CoMMpass dataset (IA19). The causal inference model for the MM Digital Twin was constructed using Aitia's REFS™ platform (Figure 1), then *in-silico* loss-of-function experiments were performed through counterfactual simulations of all gene expressions to assess their causal effects on patient overall survival (OS) and identify the OS driver genes.

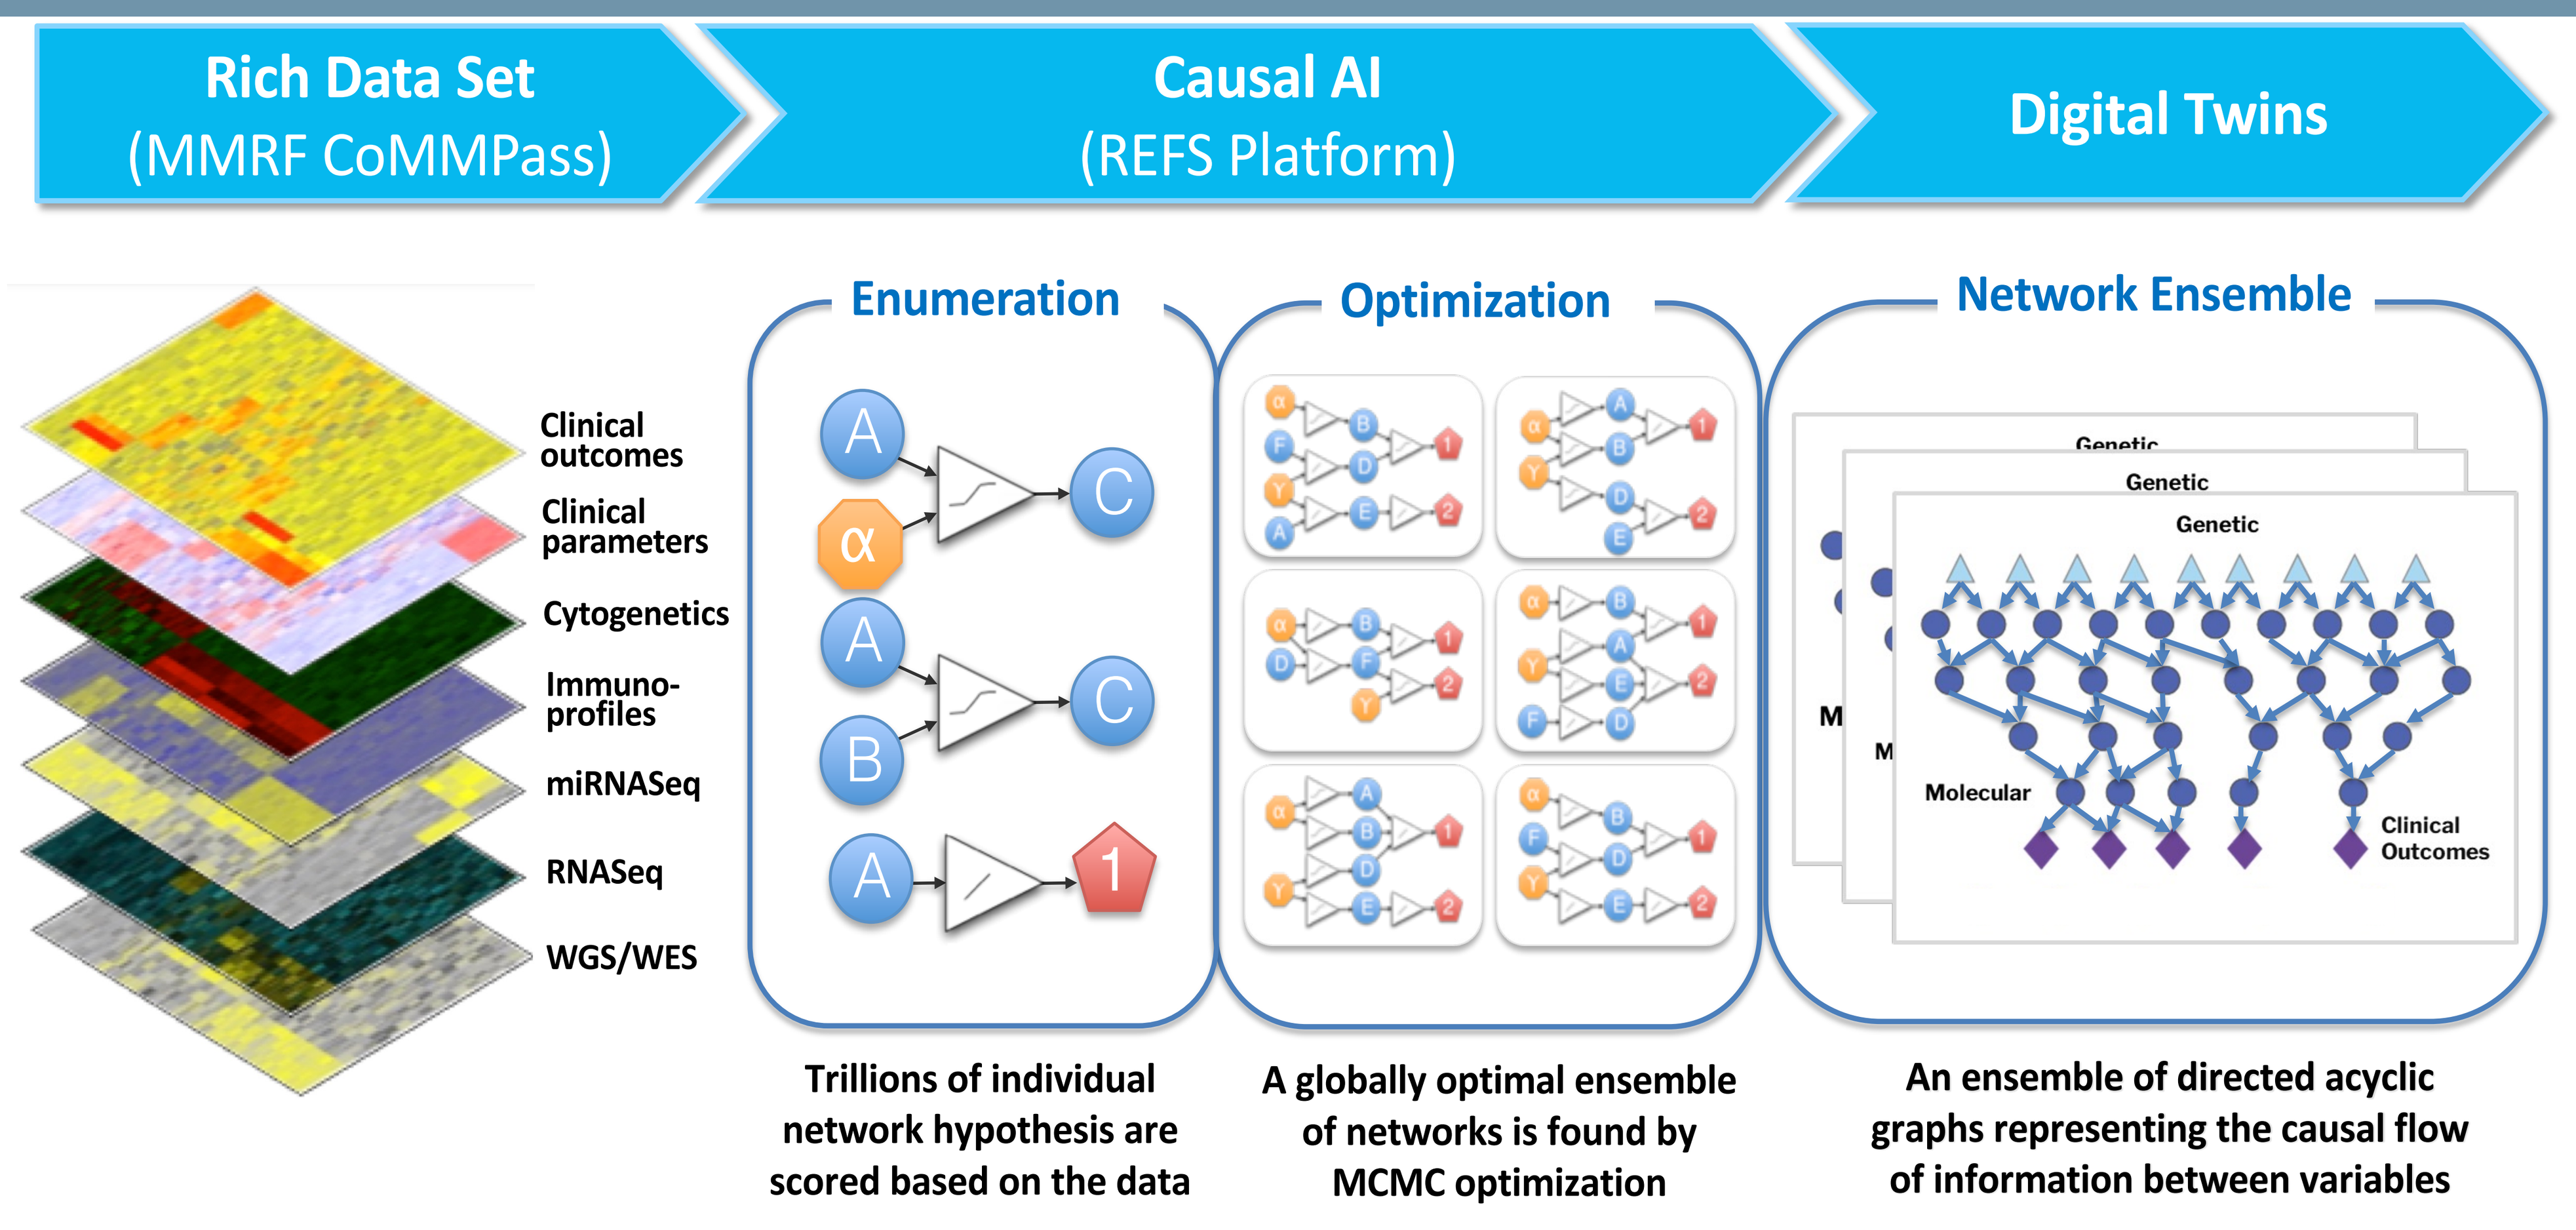

Rich Data Set
(MMRF CoMMPass)
Causal AI
(REFS Platform)
Digital Twins
Clinical outcomes
Clinical parameters
Cytogenetics
Immuno-profiles
miRNASeq
RNASeq
WGS/WES
Enumeration
Optimization
Network Ensemble
Genetic
Molecular
Clinical Outcomes
Trillions of individual network hypothesis are scored based on the data
A globally optimal ensemble of networks is found by MCMC optimization
An ensemble of directed acyclic graphs representing the causal flow of information between variables

Figure 1: Machine Learning Methods (REFS™): REFS™ workflow used to build the MM in-silico Digital Twins. After filtering for data quality and availability, we included 516 patients in the model, with 60% being hyperdiploid.

Dependency and Functional Analysis

The OS drivers were annotated using the DepMap (v.22Q4) MM cell lines data as follows:

- **MM-Dependent**: genes required for MM cell line survival
- **Non-MM Dependent**: genes required for some cancer cell line survival but not MM
- **Non-Dependent:** genes not found to be required for cancer cell line survival

Downstream genes of OS drivers were analyzed for enriched functional pathways using Ingenuity Pathway Analysis tool (IPA).

CONCLUSIONS

In this study, we demonstrate Aitia Digital Twins model can accurately screen for cancer genetic vulnerabilities in multiple myeloma and reveal their associated biomarker as well as their potential functional mechanisms. The Aitia Digital Twin model is a powerful tool in target discovery and biomarker identification and can be readily applied to other cancer types.

REFERENCES/CONTACT

1. Tsherniak A, et al. Defining a Cancer Dependency Map. Cell. 2017 Jul 27;170(3):564-576.e16.
2. Ren Z, et al. PHF19 promotes multiple myeloma tumorigenicity through PRC2 activation and broad H3K27me3 domain formation. Blood. 2019 Oct 3;134(14):1176-1189.
3. Jain P, et al. PHF19 mediated regulation of proliferation and invasiveness in prostate cancer cells. Elife. 2020 Mar 10;9:e51373.