Prediction of Amyloid PET positivity from blood-based biomarkers and clinical data using AI-based Digital Twins

Wenjun Zhu[1], So-Youn Shin[1], Jeanne Latourelle[1]

[1] Aitia, Somerville, MA, USA

Objective

- To identify individuals positive for amyloid PET results in a more cost-effective and convenient manner, accessible to a larger at-risk population
- To understand the contribution of different sources of data to the prediction of PET positivity and evaluate prediction performance across different racial & ethnic groups and disease stages

Data and Methods

Figure 1. Workflow of the Study Design

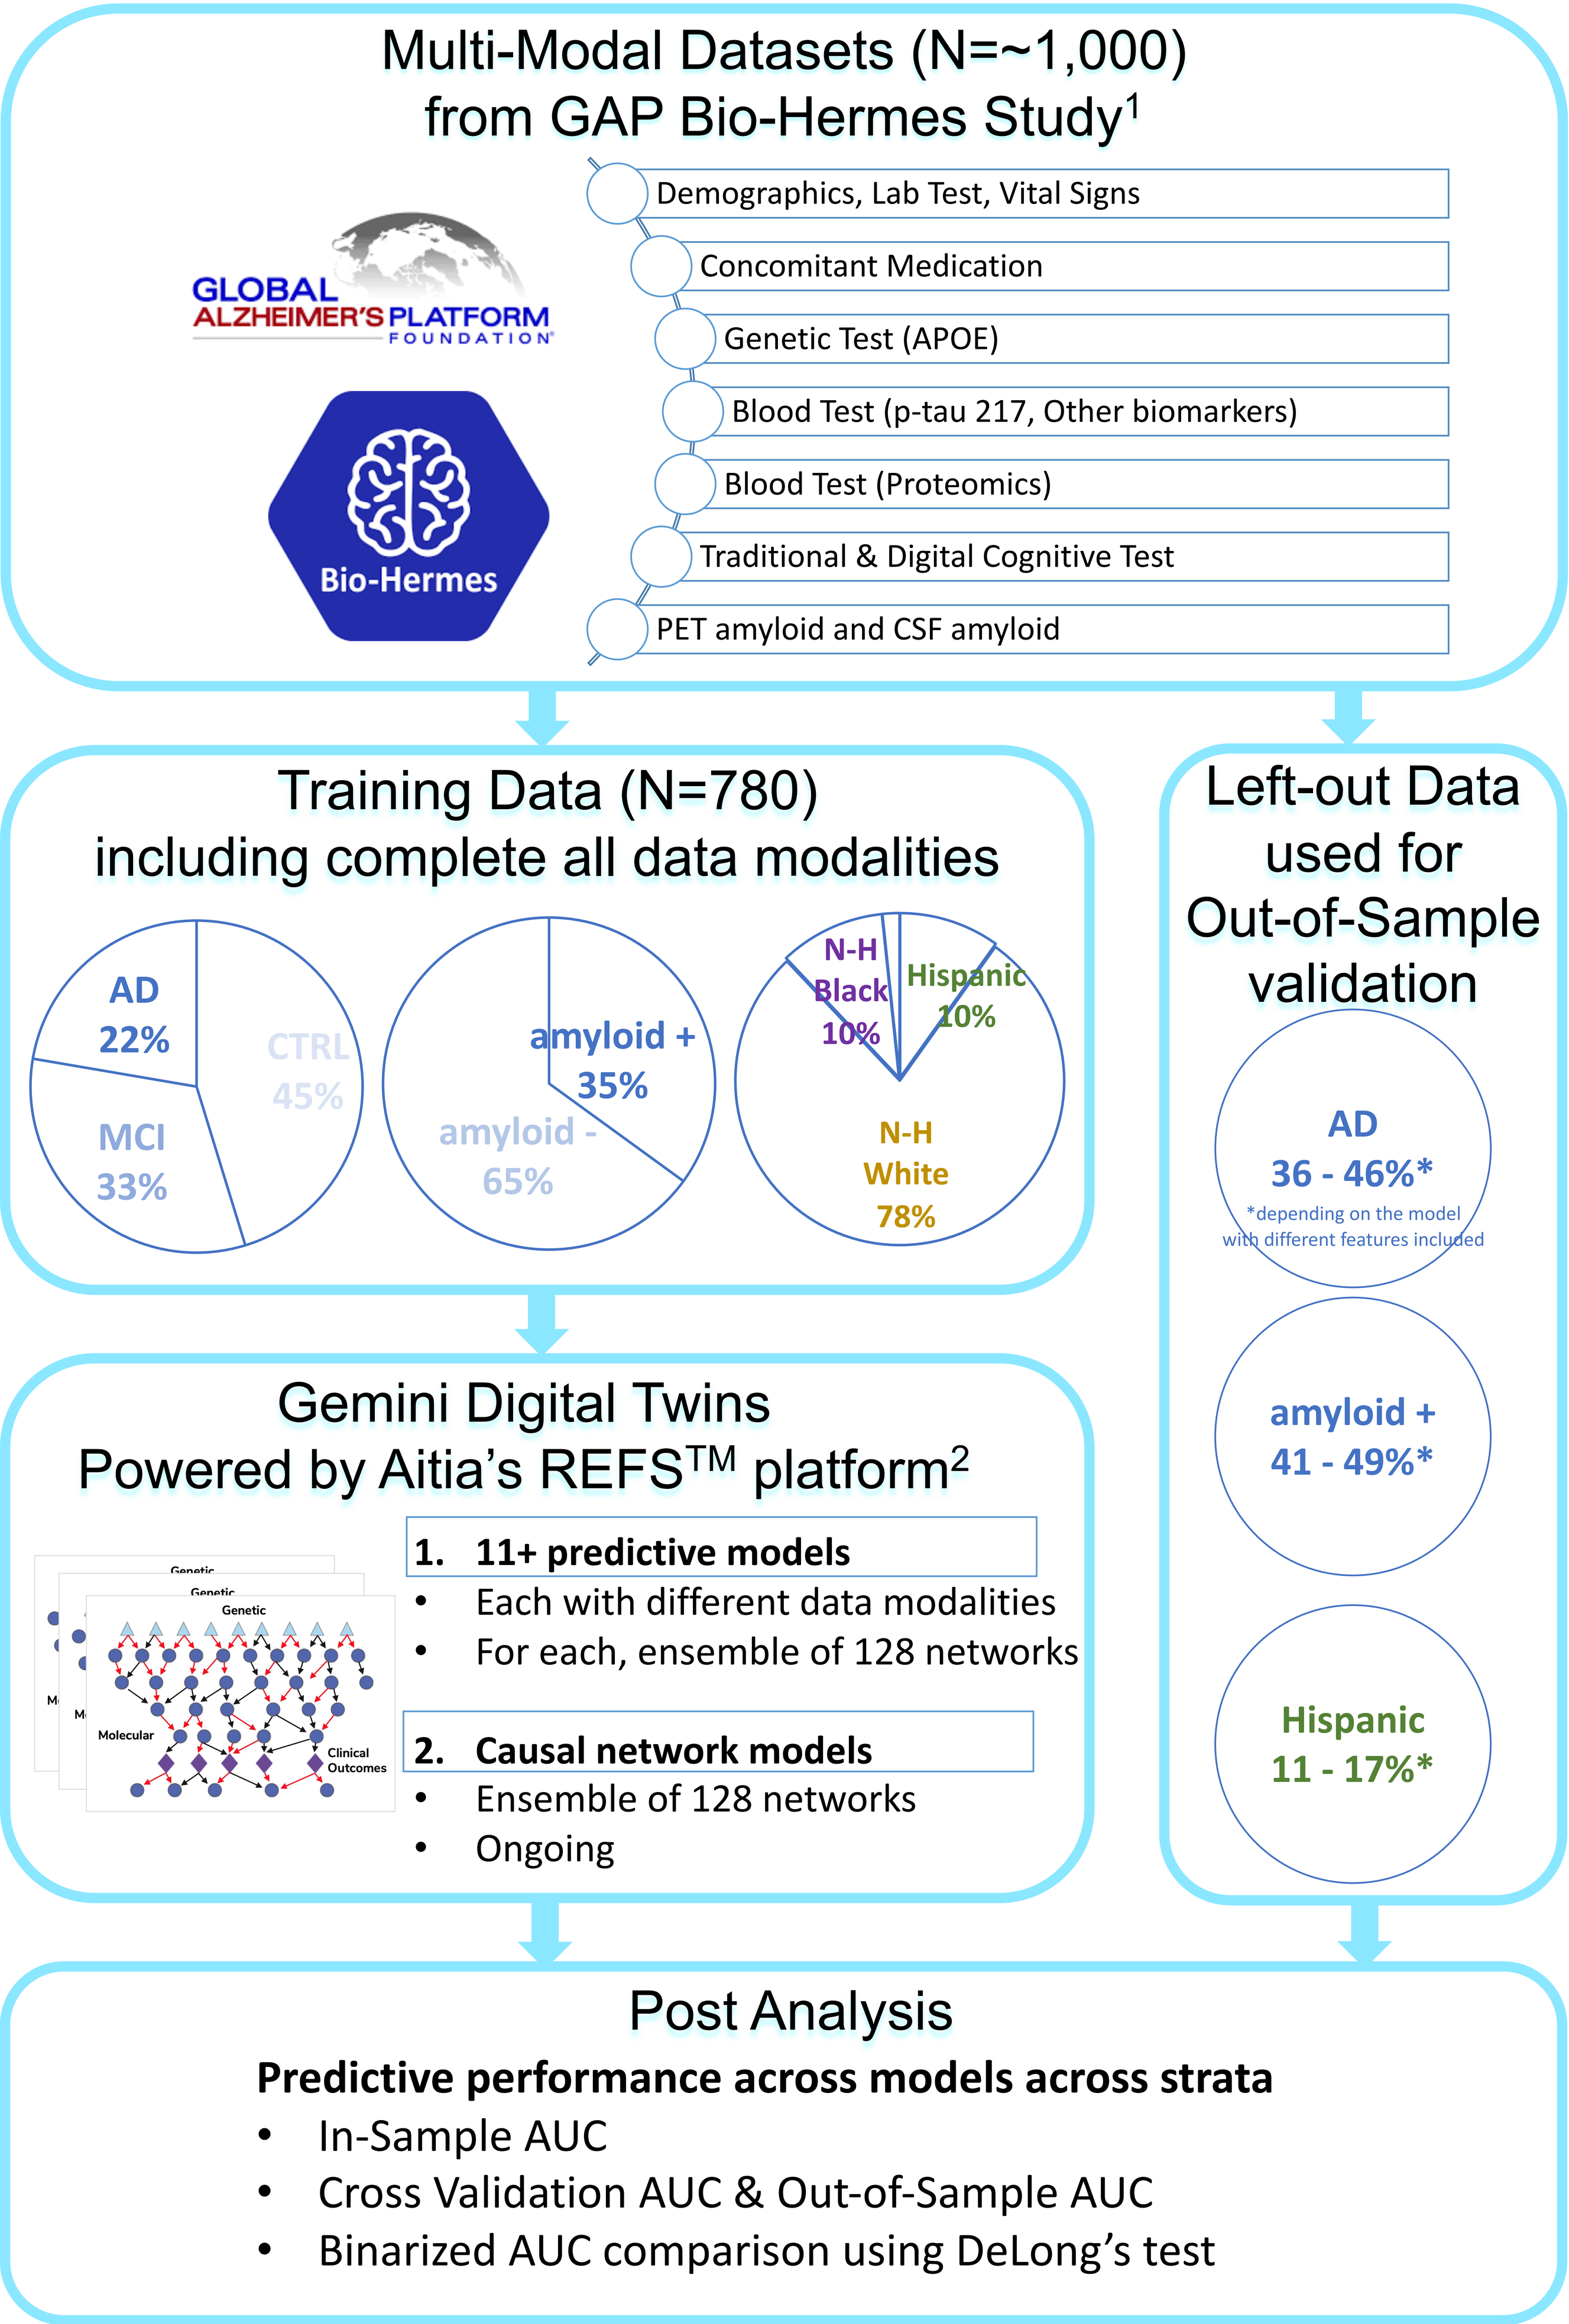
Multi-Modal Datasets (N=~1,000)
from GAP Bio-Hermes Study1
GLOBAL ALZHEIMER'S PLATFORM FOUNDATION
Bio-Hermes
Demographics, Lab Test, Vital Signs
Concomitant Medication
Genetic Test (APOE)
Blood Test (p-tau 217, Other biomarkers)
Blood Test (Proteomics)
Traditional & Digital Cognitive Test
PET amyloid and CSF amyloid
Training Data (N=780)
including complete all data modalities
AD 22%
CTRL 45%
MCI 33%
amyloid + 35%
amyloid - 65%
N-H Black 10%
Hispanic 10%
N-H White 78%
Left-out Data used for Out-of-Sample validation
AD 36 - 46%*
*depending on the model with different features included
amyloid + 41 - 49%*
Hispanic 11 - 17%*
Gemini Digital Twins
Powered by Aitia's REFSTM platform2
Genetic
Molecular
Clinical Outcomes
1. 11+ predictive models
Each with different data modalities
For each, ensemble of 128 networks
2. Causal network models
Ensemble of 128 networks
Ongoing
Post Analysis
Predictive performance across models across strata
In-Sample AUC
Cross Validation AUC & Out-of-Sample AUC
Binarized AUC comparison using DeLong's test

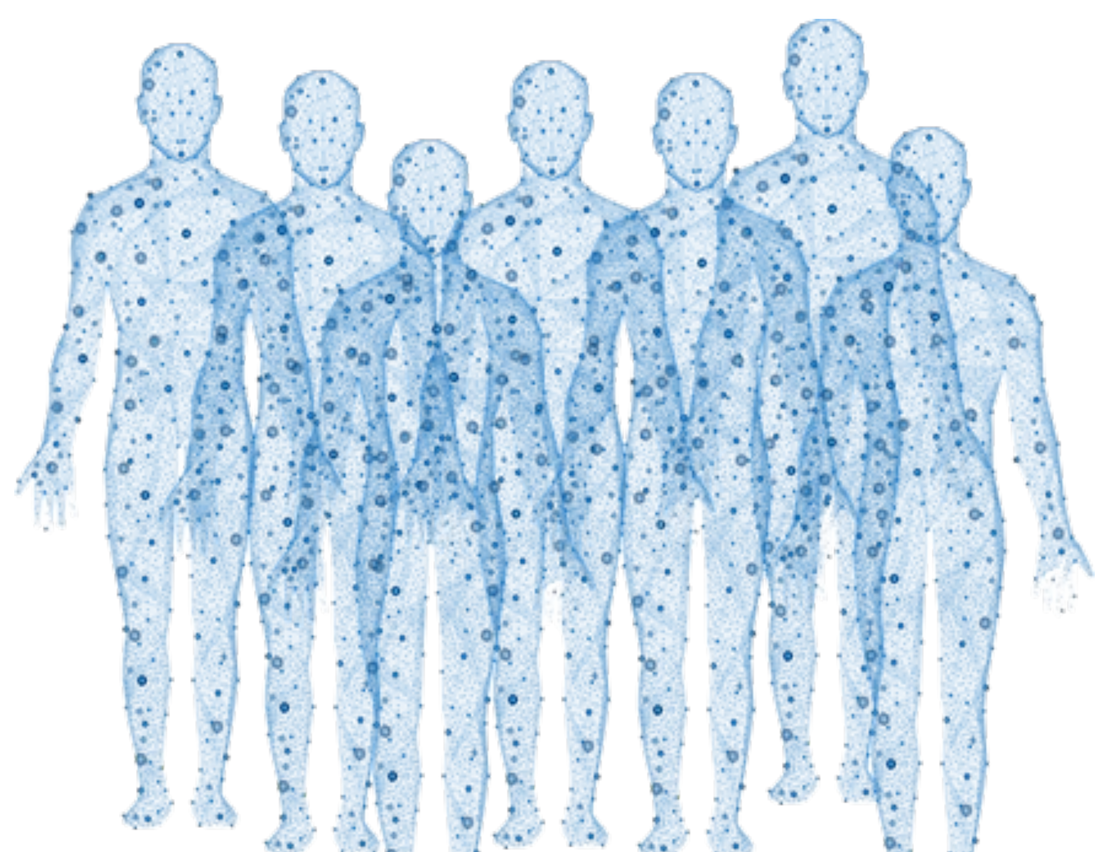

Gemini Digital Twins

- 'Gemini Digital Twins' are virtual patients modeled and simulated using the REFS™ AI platform[2] as follows.

Reverse Engineering

- Each Gemini Digital Twins is comprised of a total of 128 Bayesian network models (called an ensemble) built from the training data.
- A Bayesian network model is a directed graphical representation of relationships between variables where each node denotes a variable, and each arrow denotes a conditional dependency.

Forward Simulation

- Patient-level outcome values can be simulated in the Gemini Digital Twins, by *in silico* counterfactual experiments which computationally estimate the outcome values through model interventions, known in causal inference as 'Do' operations.
- These estimations are done fully adjusting for any confounding effects identified in the causal models, which is necessary in causal inference as emphasized in randomized experiments.

Results

Table 1. Predictive Performance of 11 models built from different data sources were compared using DeLong's Test of significant AUC Difference (p<0.05) to group into tiers of models with similar performance.

Model** (**including age, sex, race, ethnicity as predictors by default)	AUC	Model Tier defined by DeLong's Test (p>0.05) • Key Takeaways from Each Tier
All data w/ p-tau 217	0.936	**Tier 1**: Highest performance • Models including most or all key molecular markers performed equivalently to models using all available data • p-tau 217, the strongest individual marker, is able to predict Amyloid+ as well as complete data • Combining other blood biomarker data (amyloid ratio, p-tau 181, etc.), recovers all of predictive power of p-tau217, as models excluding it perform equivalently to model with it
All data w/o p-tau 217	0.933	
Biomarkers w/ p-tau 217	0.936	
Biomarkers w/o p-tau 217	0.895	
p-tau217	0.911	
Proteomics	0.838	**Tier 2**: Slightly reduced performance • Predicting Amyloid+ using only either proteomics or APOE4 genotyping also demonstrated strong performance
APOE4	0.797	
Self-reported clinical	0.71	**Tier 3**: Lower performance suitable for pre-screening • Various combinations of non-molecular clinical and cognitive measures predicted the outcome with a lower level of accuracy than the molecular features • The set of "easily accessible" self-reported clinical- and digitally assessed cognitive test data was equivalent to the traditional cognitive test • Easily accessible measurement set may have utility as more broadly implemented pre-screening tool
Digitally assessed cognitive test	0.681	
Traditionally assessed cognitive test	0.752	
EasilyAccessible Self-reported clinical + Digitally assessed cognitive test	0.72	

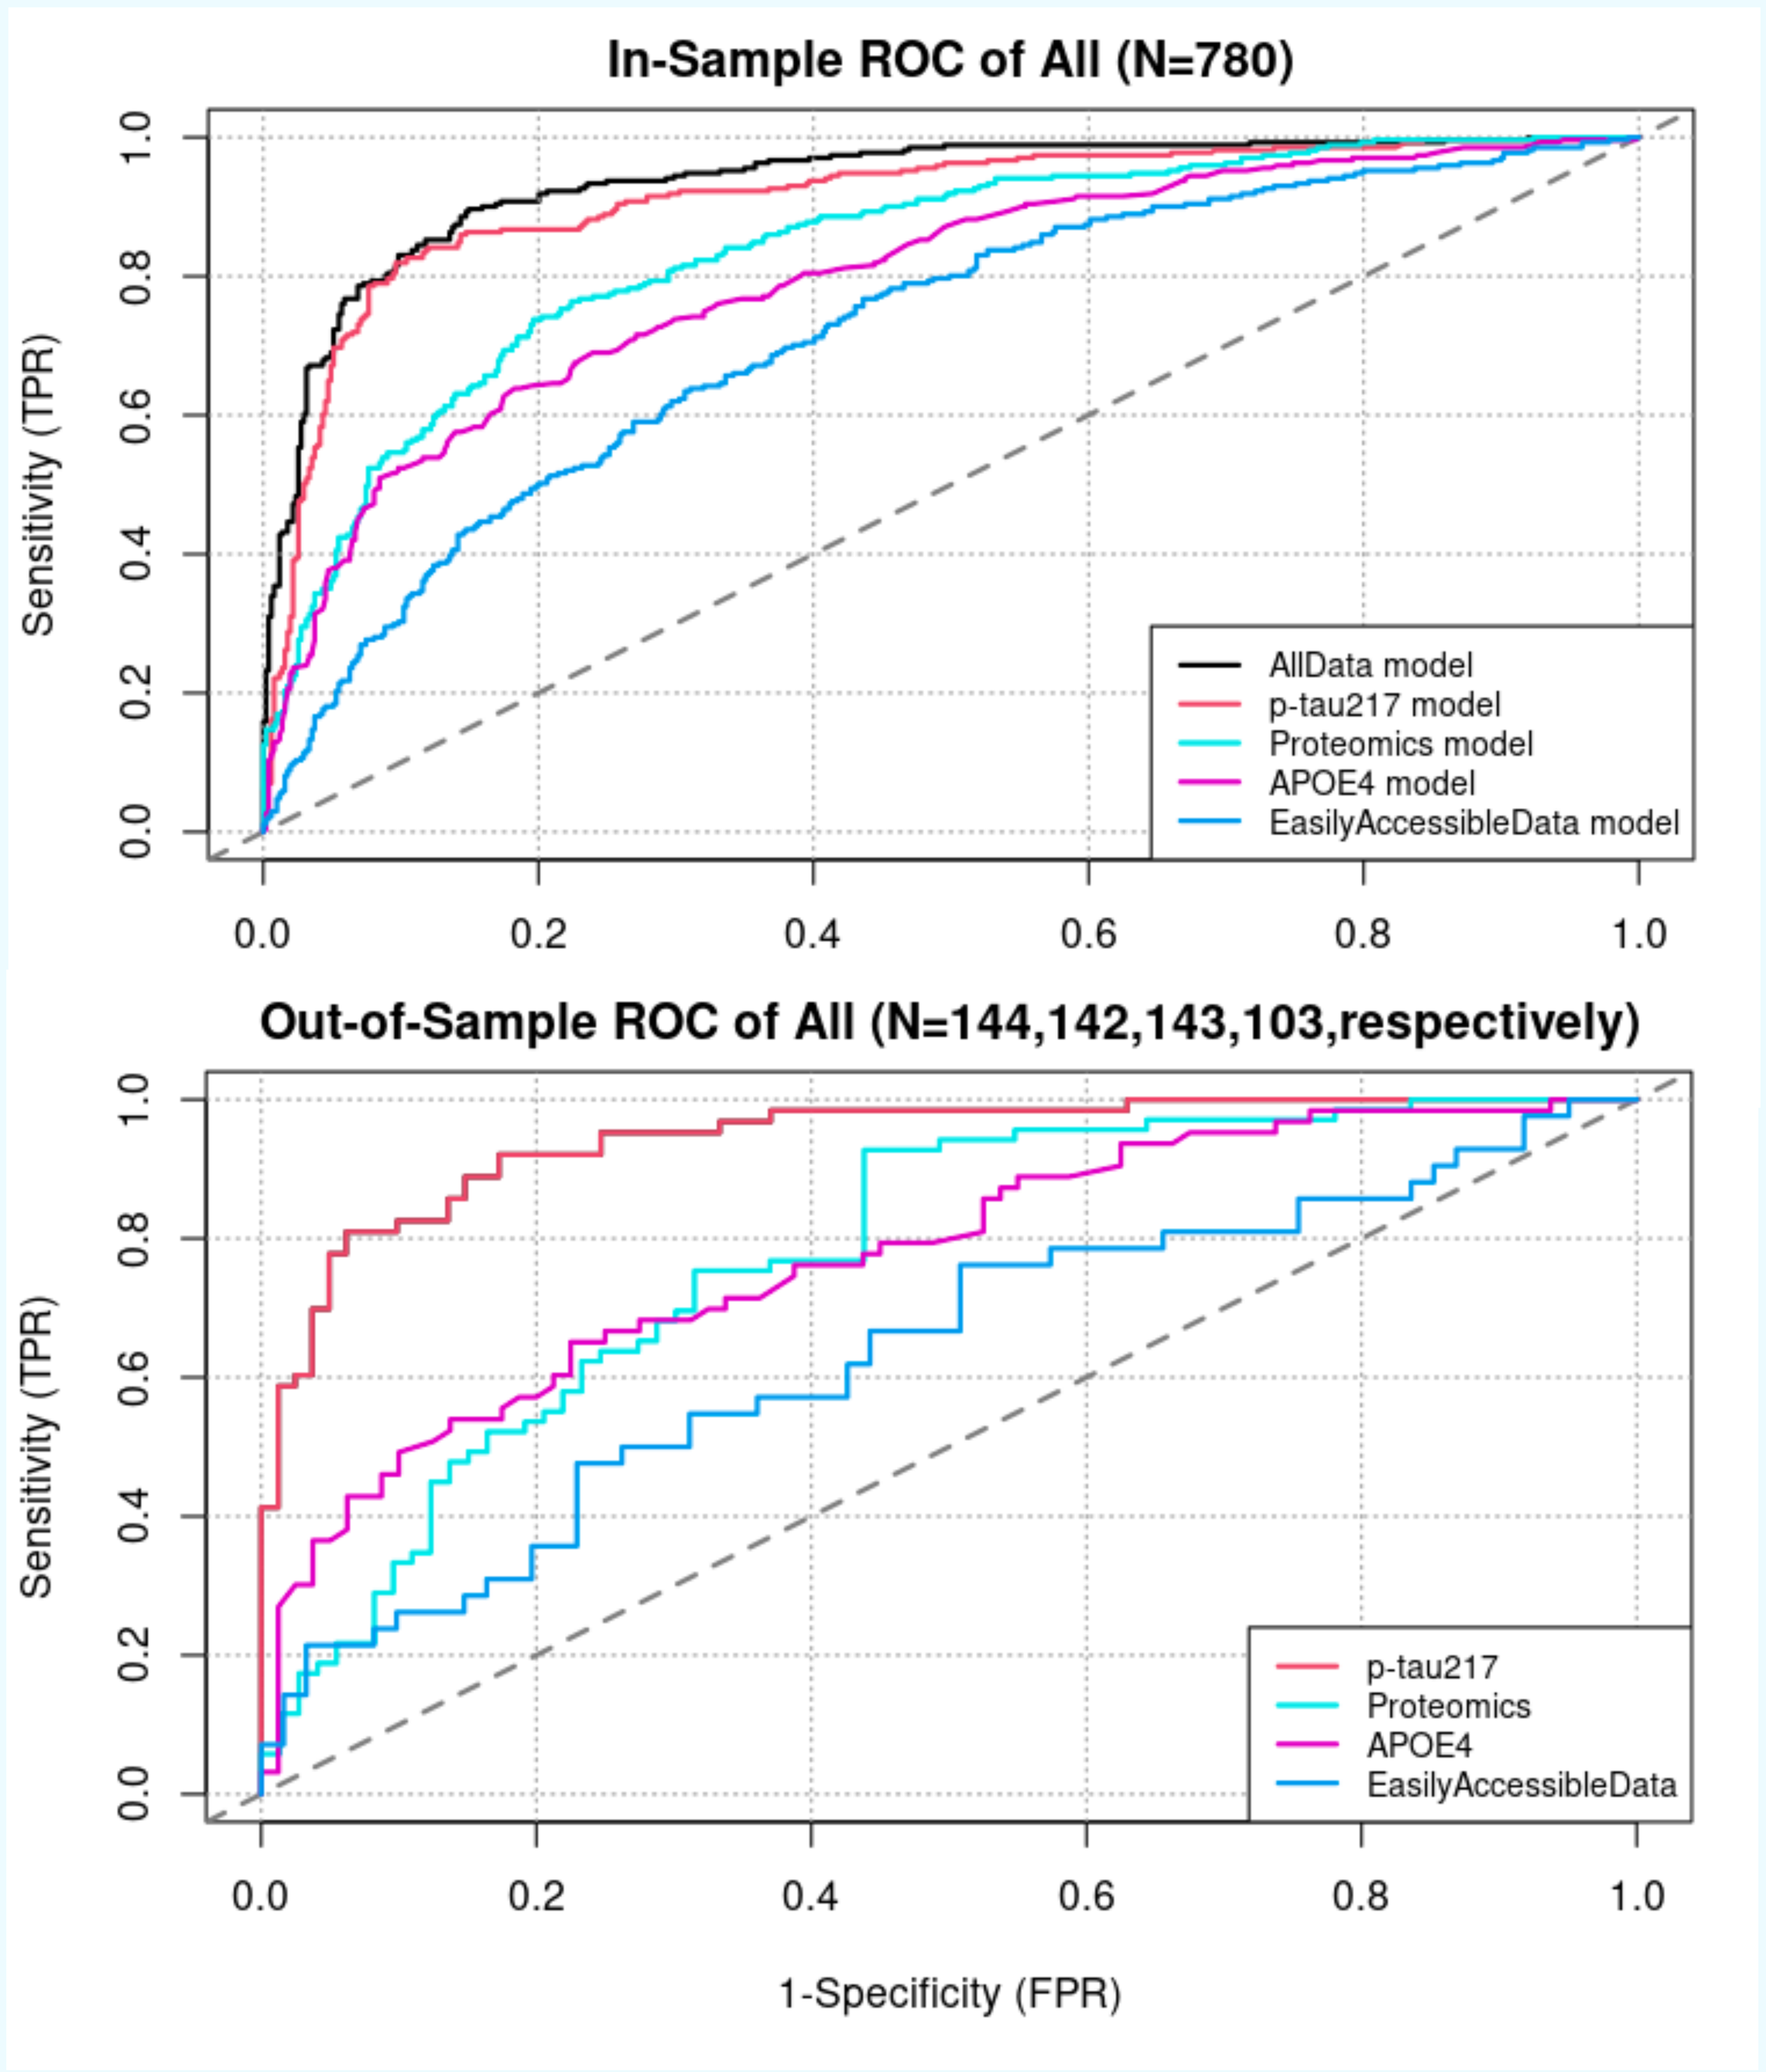
In-Sample ROC of All (N=780)
Sensitivity (TPR)
AllData model
p-tau217 model
Proteomics model
APOE4 model
EasilyAccessibleData model
Out-of-Sample ROC of All (N=144,142,143,103,respectively)
p-tau217
Proteomics
APOE4
EasilyAccessibleData
1-Specificity (FPR)

Figure 2. Representative models were tested in independent left-out cohorts and showed similar trends in performance.

Figure 3. Performance across ethnoracial groups

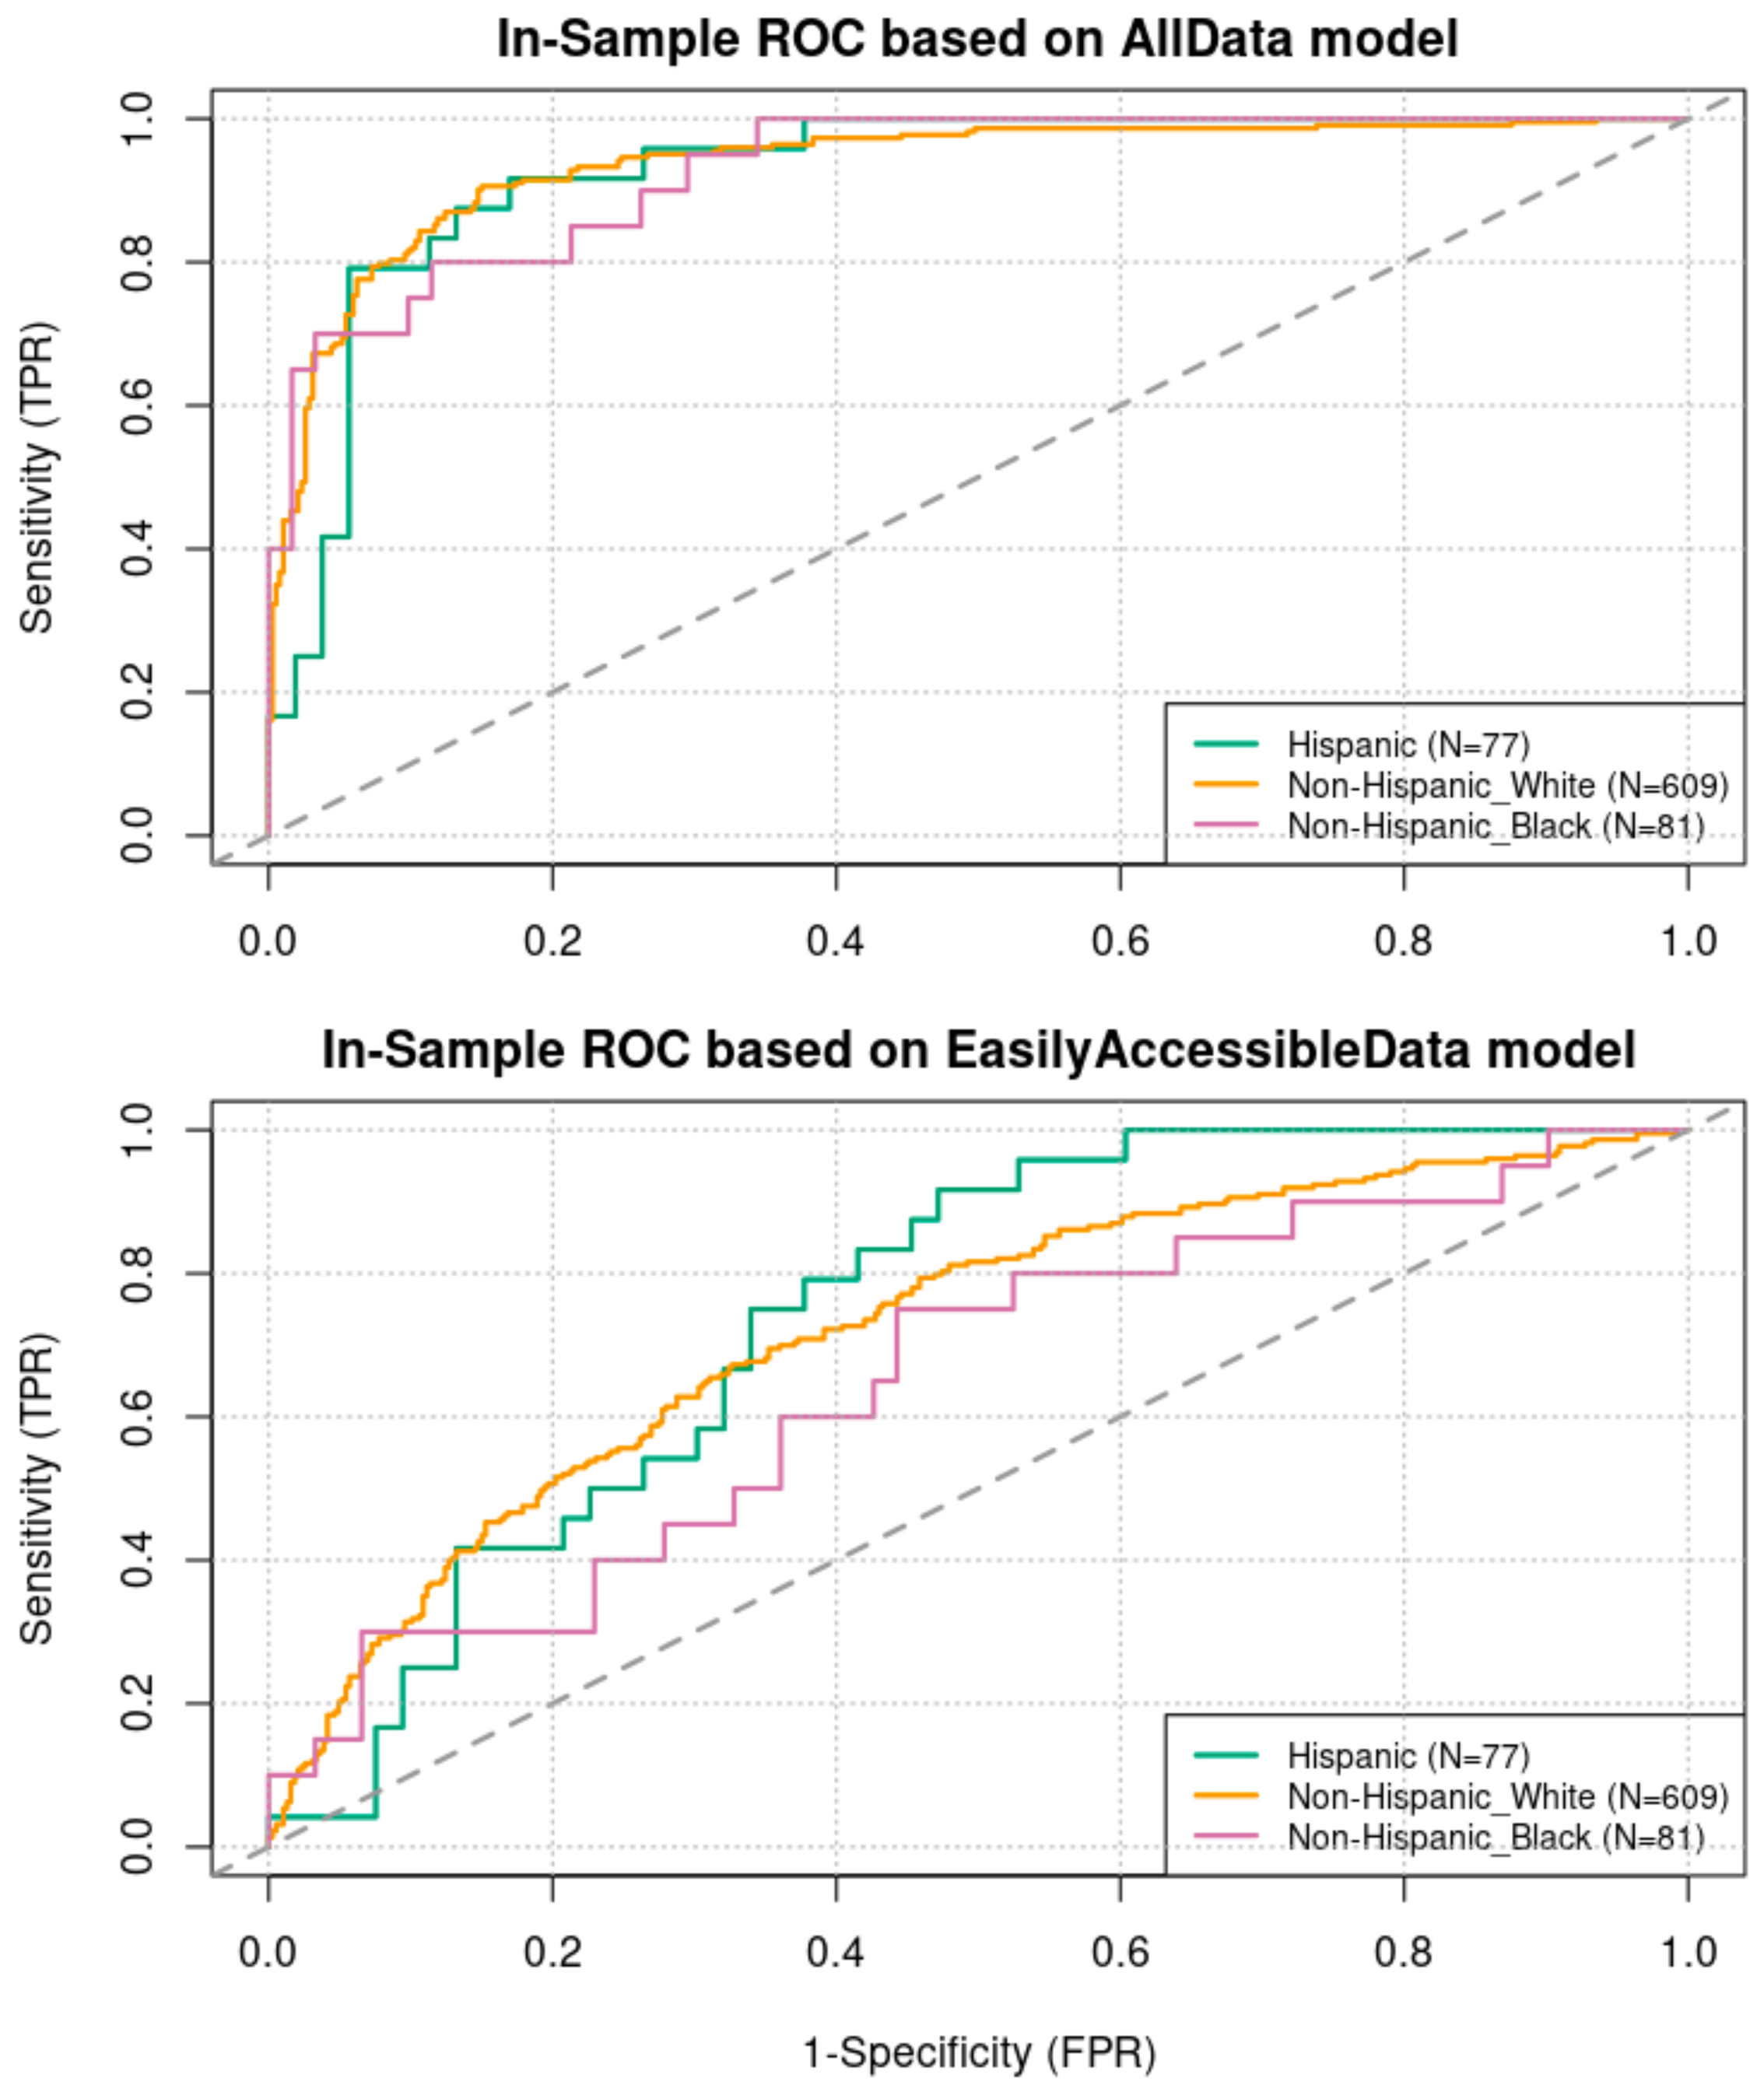
In-Sample ROC based on AllData model
Sensitivity (TPR)
Hispanic (N=77)
Non-Hispanic_White (N=609)
Non-Hispanic_Black (N=81)
In-Sample ROC based on EasilyAccessibleData model
1-Specificity (FPR)

- Performance of the models was consistent across different ethnic and racial groups for all data sources (All Data and Easily Accessible data models shown in Figure 2).
- Performance was consistent in 5-fold cross validation where median AUCs were within <1% of in-sample demonstrating robustness of models.

Conclusions

- Digital Twins showed that a blood-based biomarker, p-tau 217, is the most effective biomarker in general, corroborating previous studies.
- Digital Twins can maintain equivalent predictive performance with other blood biomarkers even in the absence of p-tau 217 and can allow utilization of easily accessible and self-reported data in a cost-effective screening approach.
- When powered by sufficient data, as collected in Bio-Hermes study, Digital Twins are able to generate equally accurate predictions in under-represented populations.
- Digital Twins can be further explored for causal inference including understanding the causal relationships among biomarkers, proteins, and disease features, potentially allowing rich understanding of biomarkers, surrogate endpoints, and disease mechanisms.

References

1. https://globalalzplatform.org/biohermesstudy/
2. Latourelle JC, et al. Lancet Neurol. 2017. PMID: 28958801. https://doi.org/10.1016/S1474-4422(17)30328-9

LP053